The novel anxiolytic compound BNC210 is a negative allosteric modulator of the alpha 7 nicotinic acetylcholine receptor

S.M. O'Connor [1], A.A. Grishin[1], E. Poiraud[2], B. Huyard[2], C. Coles[1], Y. Kolev[1], S. Wagner[2], E. Andriambeloson[2]

1. **Bionomics**, 31 Dalgleish Street, Thebarton SA 5031, Australia 2. **Neurofit SAS**, Parc d'Innovation, Boulevard Sebastien Brant, Illkirch, 67400, France

Bionomics

NEUROFIT PRECLINICAL RESEARCH

NEUROSCIENCE 2014

Y13 / 321.25

INTRODUCTION

BNC210 is under development for the treatment of anxiety disorders. Preclinical investigations showed BNC210 had anxiolytic efficacy similar to Diazepam without benzodiazepine-like side effects. This was further demonstrated in a Phase 1b study where, unlike Lorazepam, BNC210 did not cause cognitive impairment, sedation, coordination deficits or produce feelings associated with drugs of addiction. In rats, BNC210 was able to reverse the anxiety produced by CCK-8 and CCK-4 peptides. It was subsequently tested in the CCK-4 induced panic model in healthy volunteers, and significantly reduced the number and intensity of panic symptoms, and ACTH levels. BNC210 has been evaluated in 148 healthy volunteers in 5 Phase I studies to date and is safe and well tolerated.

In vitro studies have shown that BNC210 is an antagonist of the α7 nicotinic acetylcholine receptor (α7 nAChR). It inhibits rat and human α7 nAChR currents (in stably transfected cell lines) induced by acetylcholine, nicotine, choline and the α7 specific agonist PNU-282987, with IC_{50} values in the range of 1.2-3μM. BNC210 does not displace alpha-bungarotoxin binding and its inhibitory effects are not influenced by the concentration of acetylcholine used (EC_{20} or EC_{80}), suggesting that the modulation is via an allosteric site.

Although α7 nAChR antagonists have been associated with antidepressant activity, this target is novel for anxiolytics. We sought to demonstrate that the in vivo anxiolytic activity of BNC210 is achieved through its negative allosteric modulation (NAM) of α7 nAChR. The α7 specific agonist PNU-282987 displays an anxiogenic effect in the rat Open Field (1). This compound was selected for its potential to block the anxiolytic activity of BNC210 in vivo. The rat elevated plus maze (EPM) was the model used to demonstrate the anxiogenic effect of PNU-282987, and then reversal of this effect by the α7 nAChR specific antagonists, methyllycaconitine (MLA, competitive) and BNC210 (non-competitive).

These data support the hypothesis that the anxiolytic effect of BNC210 is achieved through negative allosteric modulation of α7 nAChR and indicates the involvement of this target in the modulation of anxiety.

METHODS

ELECTROPHYSIOLOGY

GH4C1 cells stably expressing rat or human α7 nAChRs were patch-clamped in the recording chamber of 16-channel re-usable Dynaflow ReSolve® chip using EPC10 USB amplifier (HEKA Elektronik, Germany). Extracellular solution contained NaCl (137 mM), KCl (5 mM), $CaCl_2$ (2.5 mM), $MgCl_2$ (1 mM), HEPES (10 mM), D-Glucose (10 mM), pH7.4. Thin wall borosilicate glass electrodes (Harvard Apparatus) were pulled to a resistance of 2-4 MΩ when filled with intracellular solution (K+-gluconate (120 mM), KCl (5 mM), HEPES (10 mM), EGTA (10 mM), $MgCl_2$ (1 mM), ATP (2 mM, pH7.2)). Cells were held at -70 mV. Cells with series resistance below 15 MΩ were kept and 40% compensation was utilized routinely. Concentration-response curves for selected compounds were fitted and plotted in Prism5 (GraphPad Software, Inc., CA).

ELEVATED PLUS MAZE AND BASIC ANXIETY STATUS LEVEL

The elevated plus maze (EPM) (2) is based on the conflict between the innate tendency of rodents to explore novel environments and to avoid open, brightly lit areas and is used to evaluate relative anxiety status. The 'plus-shaped' apparatus consists of four exploratory arms (45 × 10 cm) which are interconnected by a small platform (10 × 10 cm). Two arms are well lit and open and the other two are enclosed with 30 cm high walls and dimly lit. The apparatus is placed 66 cm above the floor. In this task the rat is placed in the centre of the maze from where it can walk down any of four runways. Rats prefer the closed arms but will venture out into the open arms. The amount of time spent and entries into the open arms are recorded. Rats in "high anxiety status" spend less time and make fewer entries into the open arms compared to rats in "low anxiety status".

The anxiolytic effects of compounds are tested in rats with high basal anxiety levels. In contrast, the effect of anxiogenic compounds are investigated in rats with low basal anxiety levels. To reduce the level of basal anxiety in rats, they are handled and habituated to the experimenter on a daily basis for 7 days prior the EPM experiments. Rats with high level of basal anxiety are not subject to the pre-handling process. BNC210 (p.o.) was dosed 1 hour prior to testing and PNU-282987 and MLA (both i.p.) were dosed 40 minutes prior to testing.

RESULTS

1. THE INHIBITORY EFFECTS OF BNC210 ARE NOT INFLUENCED BY ACETYLCHOLINE CONCENTRATION SUGGESTING THAT MODULATION IS VIA AN ALLOSTERIC SITE

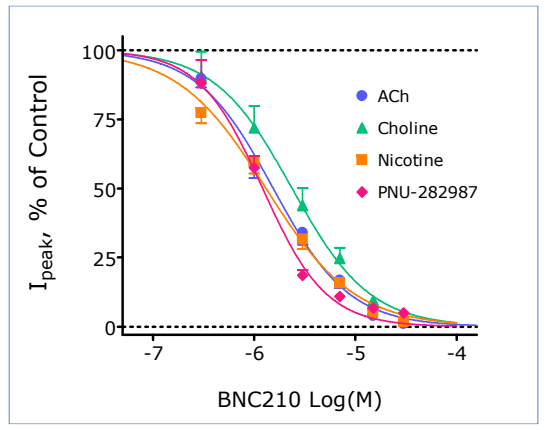
Ipeak, % of Control
ACh
Choline
Nicotine
PNU-282987
BNC210 Log(M)

IC_{50} values of BNC210 on rat α7 nAChR		
Agonist (EC_{80}):	IC_{50} (μM)	n_H
ACh	1.5	1.16
Nicotine	1.3	1.00
Choline	2.4	1.13
PNU-282987	1.2	1.38

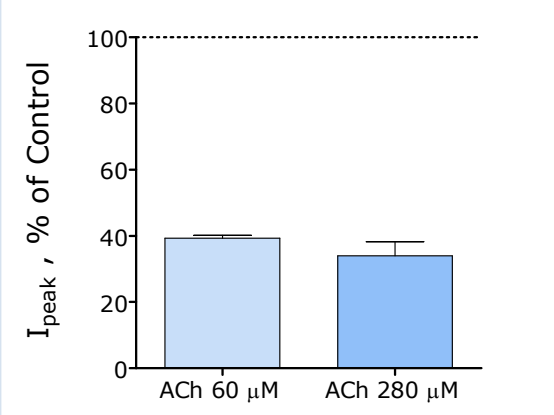
Ipeak , % of Control
ACh 60 μM
ACh 280 μM

BNC210 IC_{50} values were determined in manual patch-clamp electrophysiology on rat α7 nAChR expressed in GH4C1 cells, and obtained with EC_{80} concentrations of acetylcholine, nicotine, choline and PNU-282987. IC_{50} values of BNC210 were consistent across ligands and similar values were seen with human α7 nAChR, validating target specificity (Mean ± SEM, n=3-4). In electrophysiology, 3 μM BNC210 gave ~40% inhibition of rat α7 nAChR currents produced by ACh at an EC_{20} or EC_{80} concentration. The same degree of inhibition was produced independently of the ACh concentration suggesting that BNC210 acts non-competitively (Mean ± SEM, n=5).

2. THE ANXIOGENIC EFFECT OF PNU-282987 IS REVERSED BY METHYLLYCACONITINE

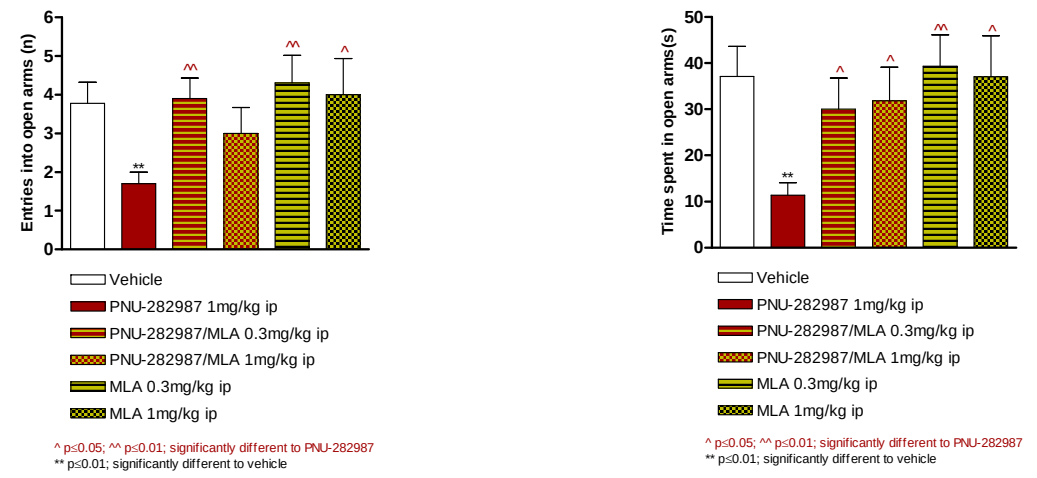
Entries into open arms (n)
Time spent in open arms(s)
Vehicle
PNU-282987 1mg/kg ip
PNU-282987/MLA 0.3mg/kg ip
PNU-282987/MLA 1mg/kg ip
MLA 0.3mg/kg ip
MLA 1mg/kg ip
^ p≤0.05; ^^ p≤0.01; significantly different to PNU-282987
** p≤0.01; significantly different to vehicle

The anxiogenic effect of PNU-282987 was tested in rats with 'low basal anxiety' (control animals- average entries ~4; average time spent ~40 s) to facilitate demonstration of the anxiogenic effect. At 1mg/kg, i.p., PNU-282987 generated anxiety as shown by significant reduction of entries into and time spent in the open arms of the EPM. MLA (0.3, 1mg/kg, i.p.) fully reversed the anxiogenic effect of PNU-282987 and restored animal behaviour to the same level as control animals for Entries and Time. MLA alone did not have an effect on animal behaviour. (Mean ± SEM, n=10).

CONCLUSIONS

- BNC210 is a negative allosteric modulator of the α7 nicotinic acetylcholine receptor.
- α7 nicotinic acetylcholine receptor is a novel target for anxiolysis.
- BNC210 inhibits rat and human α7 nAChR currents induced by acetylcholine, nicotine, choline and PNU-282987 in vitro.
- Specific α7 agonist PNU-282987 blocked the anxiolytic effect of BNC210 in vivo.
- BNC210 reversed the anxiogenic effect of specific α7 agonist PNU-282987 in vivo.
- Negative allosteric modulation of α7 nAChR is the mechanism for the anxiolytic effect of BNC210.

RESULTS

3. THE ANXIOGENIC EFFECT OF PNU-282987 IS REVERSED BY THE ANTAGONIST, BNC210

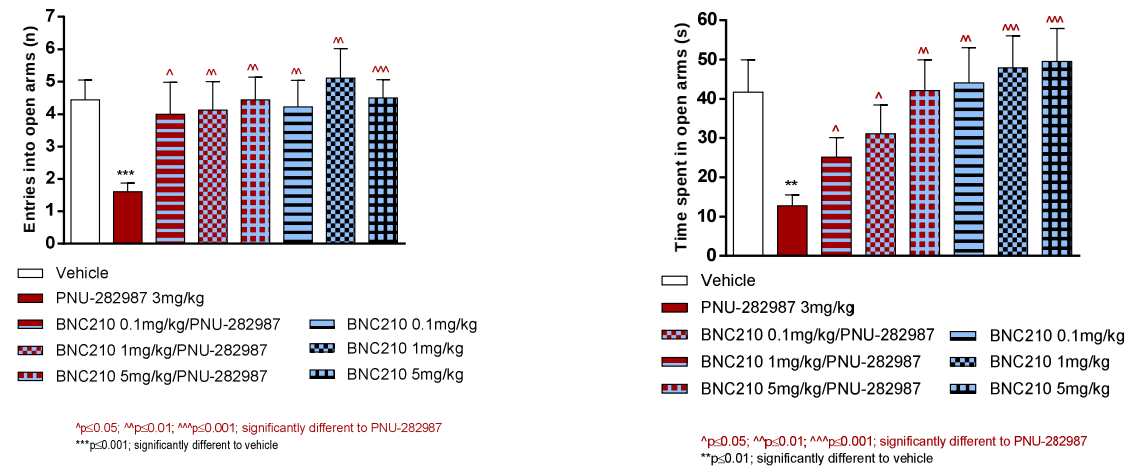
Entries into open arms (n)
Time spent in open arms (s)
Vehicle
PNU-282987 3mg/kg
BNC210 0.1mg/kg/PNU-282987
BNC210 1mg/kg/PNU-282987
BNC210 5mg/kg/PNU-282987
BNC210 0.1mg/kg
BNC210 1mg/kg
BNC210 5mg/kg
^p≤0.05; ^^p≤0.01; ^^^p≤0.001; significantly different to PNU-282987
***p≤0.001; significantly different to vehicle
**p≤0.01; significantly different to vehicle

The anxiogenic activity of PNU-282987 (3mg/kg, i.p.) was reversed by BNC210 in a dose-dependent manner in the EPM. Co-dosing with BNC210 (0.1, 1, 5mg/kg, p.o.) significantly reversed anxiogenesis by increasing entries into and time spent in the open arms of the maze. Full reversal was achieved for Entries with all doses of BNC210 whereas the effect on Time appeared to have some dose dependency. Treatment with BNC210 alone did not have an effect on rat behaviour in the 'low basal anxiety' conditions. (Mean ± SEM, n=12).

4. THE ANXIOLYTIC EFFECT OF BNC210 IS REVERSED BY THE α7 AGONIST PNU-282987

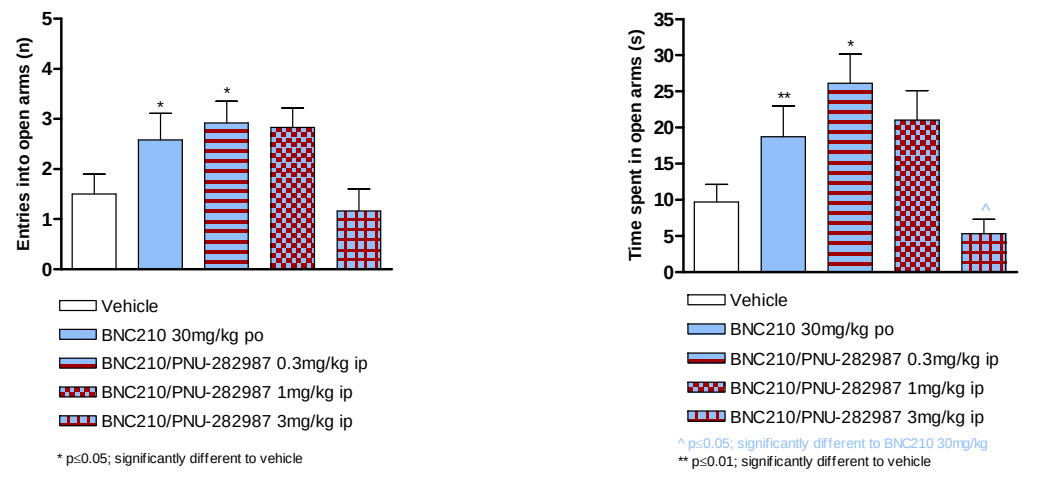
Entries into open arms (n)
Time spent in open arms (s)
Vehicle
BNC210 30mg/kg po
BNC210/PNU-282987 0.3mg/kg ip
BNC210/PNU-282987 1mg/kg ip
BNC210/PNU-282987 3mg/kg ip
* p≤0.05; significantly different to vehicle
^ p≤0.05; significantly different to BNC210 30mg/kg
** p≤0.01; significantly different to vehicle

To demonstrate the anxiolytic effect of BNC210, animals being tested in the EPM were not pre-handled in order to maintain 'high basal anxiety' levels. The absence of pre-handling led to reduction in Entries and Time parameters in the vehicle control animals (average entries ~1.5; average time spent ~10 s). BNC210 (30mg/kg, p.o.) reduced the anxiety of the animals by significantly increasing time spent and entries into the open arms of the EPM. PNU-282987 was co-dosed with BNC210 at 0.3, 1 and 3mg/kg, i.p. Lower doses of PNU-282987 (0.3, 1mg/kg, i.p) did not affect BNC210 activity but complete reversal occurred with the 3 mg/kg dose as seen by the behaviour of these animals being at the level of control animals (Mean ± SEM, n=10).

REFERENCES

1. Pandya and Yakel; (2013) Neuropharmacology; 70:35-42.
2. Pellow et al; (1985) Journal of Neuroscience Methods; 14:149-167.